EXPANSION OF CD34+/90/CXCR4+ WHARTON´S JELLY MESENCHYMAL STROMAL CELLS

Ávila-Portillo, Luz Mabel[1,2], Riveros, Angela[1], Graciano Noiver[3], Ávila Jenniffer P[1], Franco Derly[1], Ortiz Paola[1], Chaparro Mauricio[4], Estupiñan Marcela[4].

1. Stem Medicina Regenerativa. 2 Hospital Militar Central, Bogotá. 3.Universidd Militar Nueva Granada. 4 Fundación Hospital Misericordia. Calle 113. 7-45 Torre B oficina 702, dircientifica@bancodecelulas.com. Bogotá,Colombia.

INTRODUCTION

Umbilical Cord Blood (UBC) has hematopoietic Stem cells (HSCs) CD34+, these have the capacity for long term bone marrow repopulation, being easily obtained and criopreserved for their posterior use, which makes them a feasible alternative for transplant. The limited volume of obtained UCB, makes the count of HSCs to be low, having a repercussion on the probability of doing a graft on short term..

Our research is a comparison of two expansion methodos of CD34 +, CD49f +, CD90 + (static culture and bioreactor) using human recombinant cytokines (HRC), Stem cell factor (SCF), Interleukin 3 (IL-3), thrombopoietin (TPO) And ligand FT3 (FT3L)) DLK-1 Interleukin 6 (IL6), umbilical cord Wharton CD146 + mesenchymal cells encapsulated in calcium alginate as mesenchymal stem cell feeder which compared to previously established models.

MATERIALS AND METHODS

Isolation CD34+ from UCB

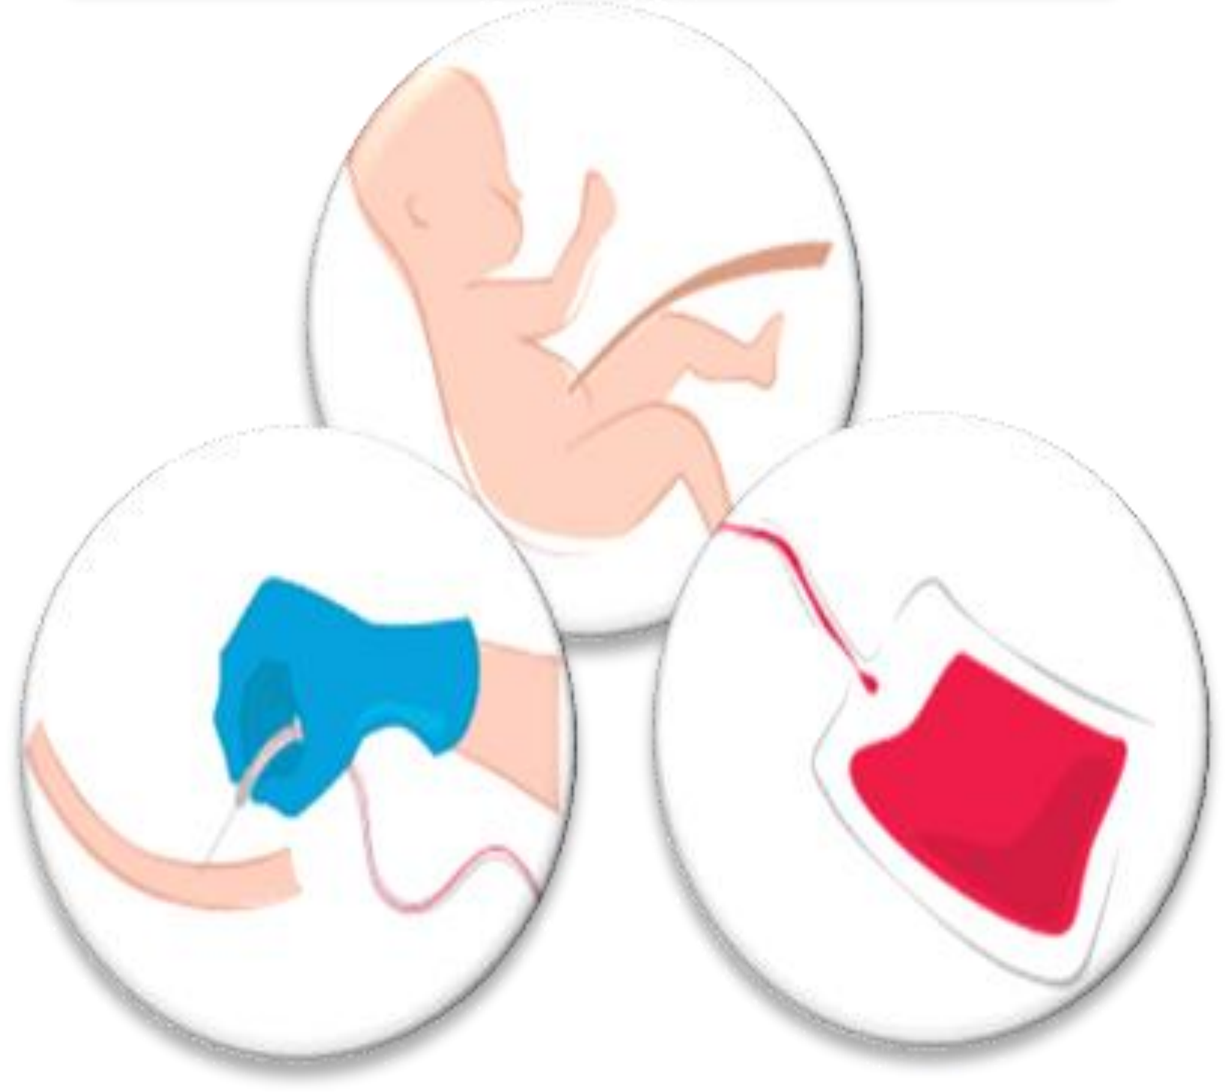

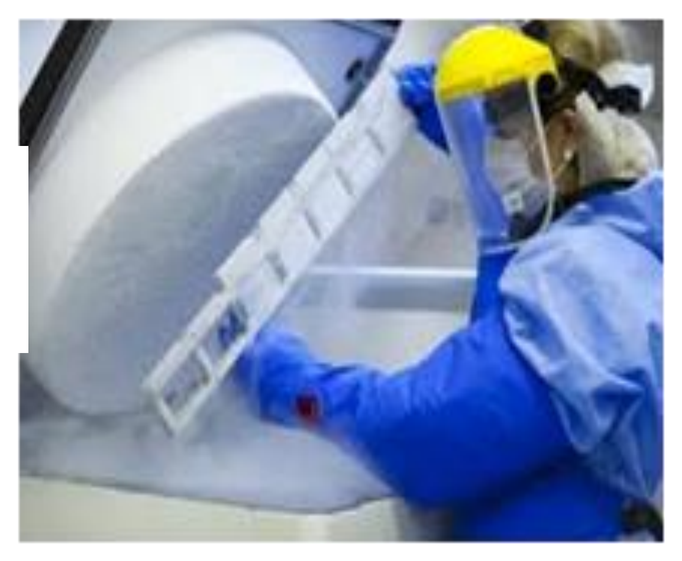

Cryopreservation and thawing

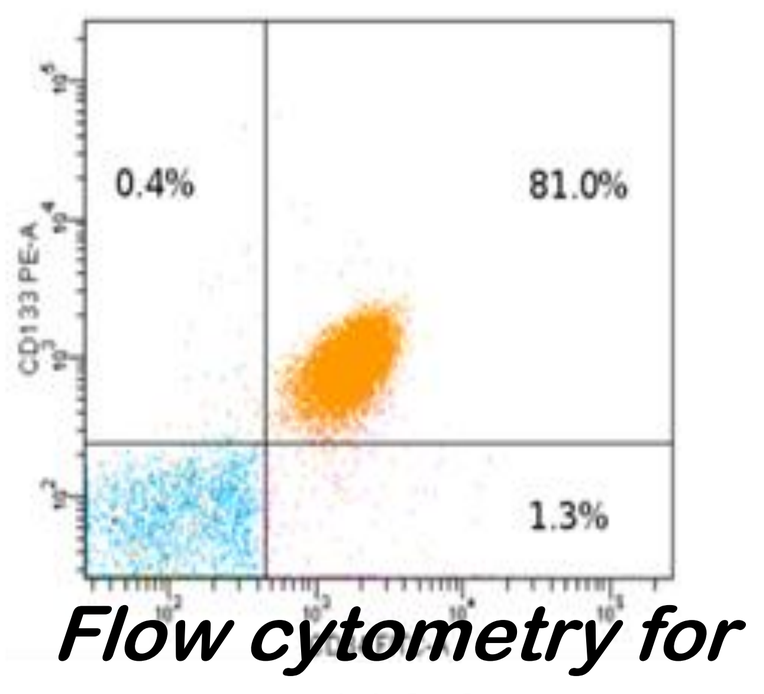
0.4%
81.0%
1.3%

Flow cytometry for HSC CD34+CD90+CD49f+

Isolation MSC from UC

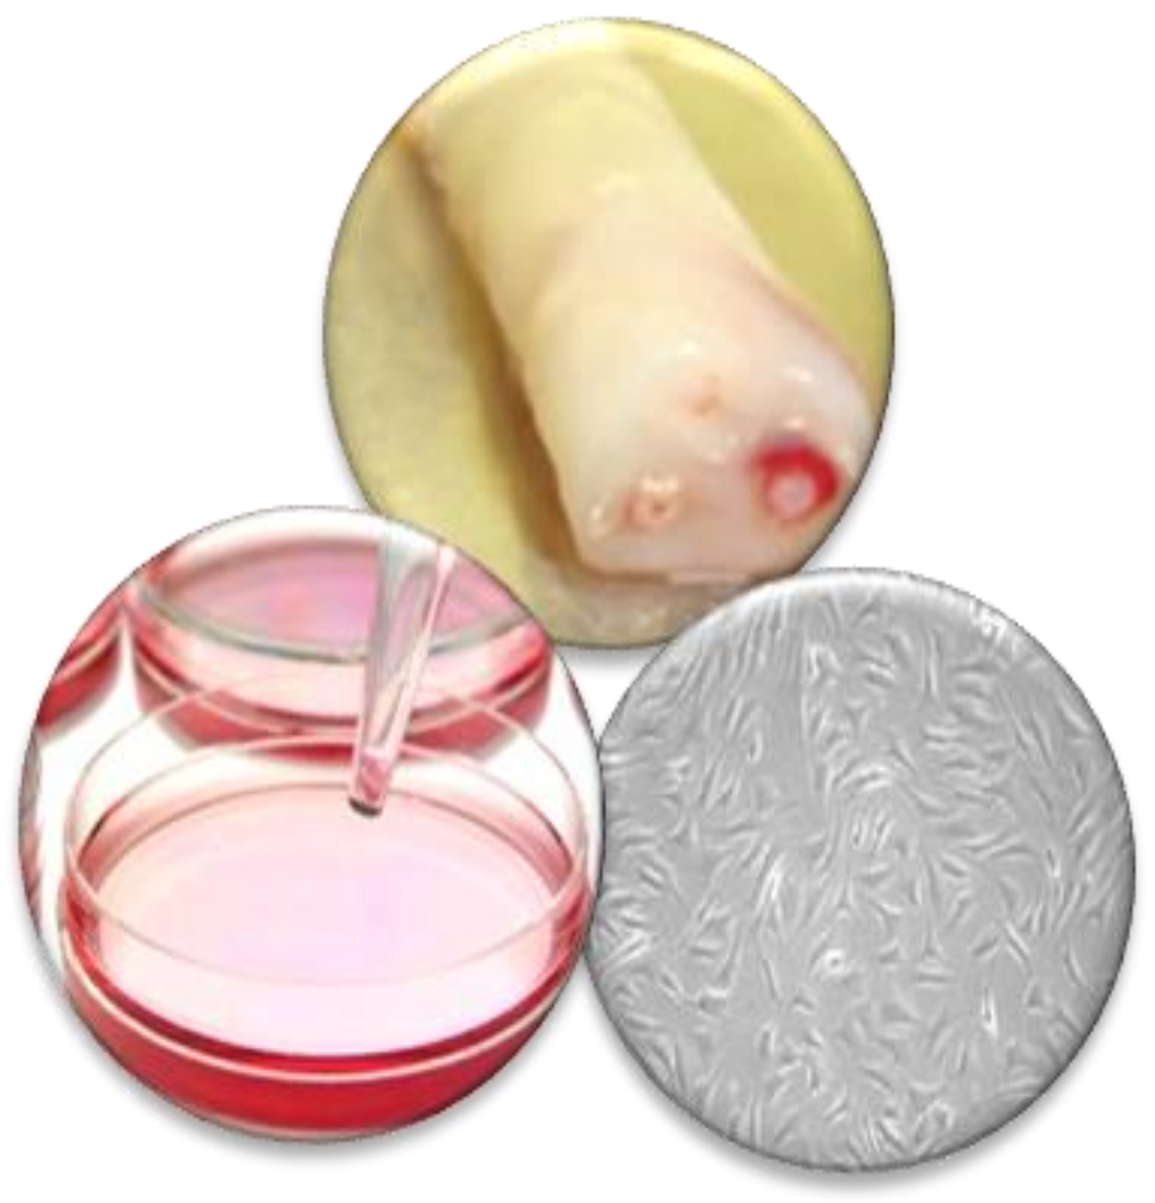

Perfusion culture *Static culture*

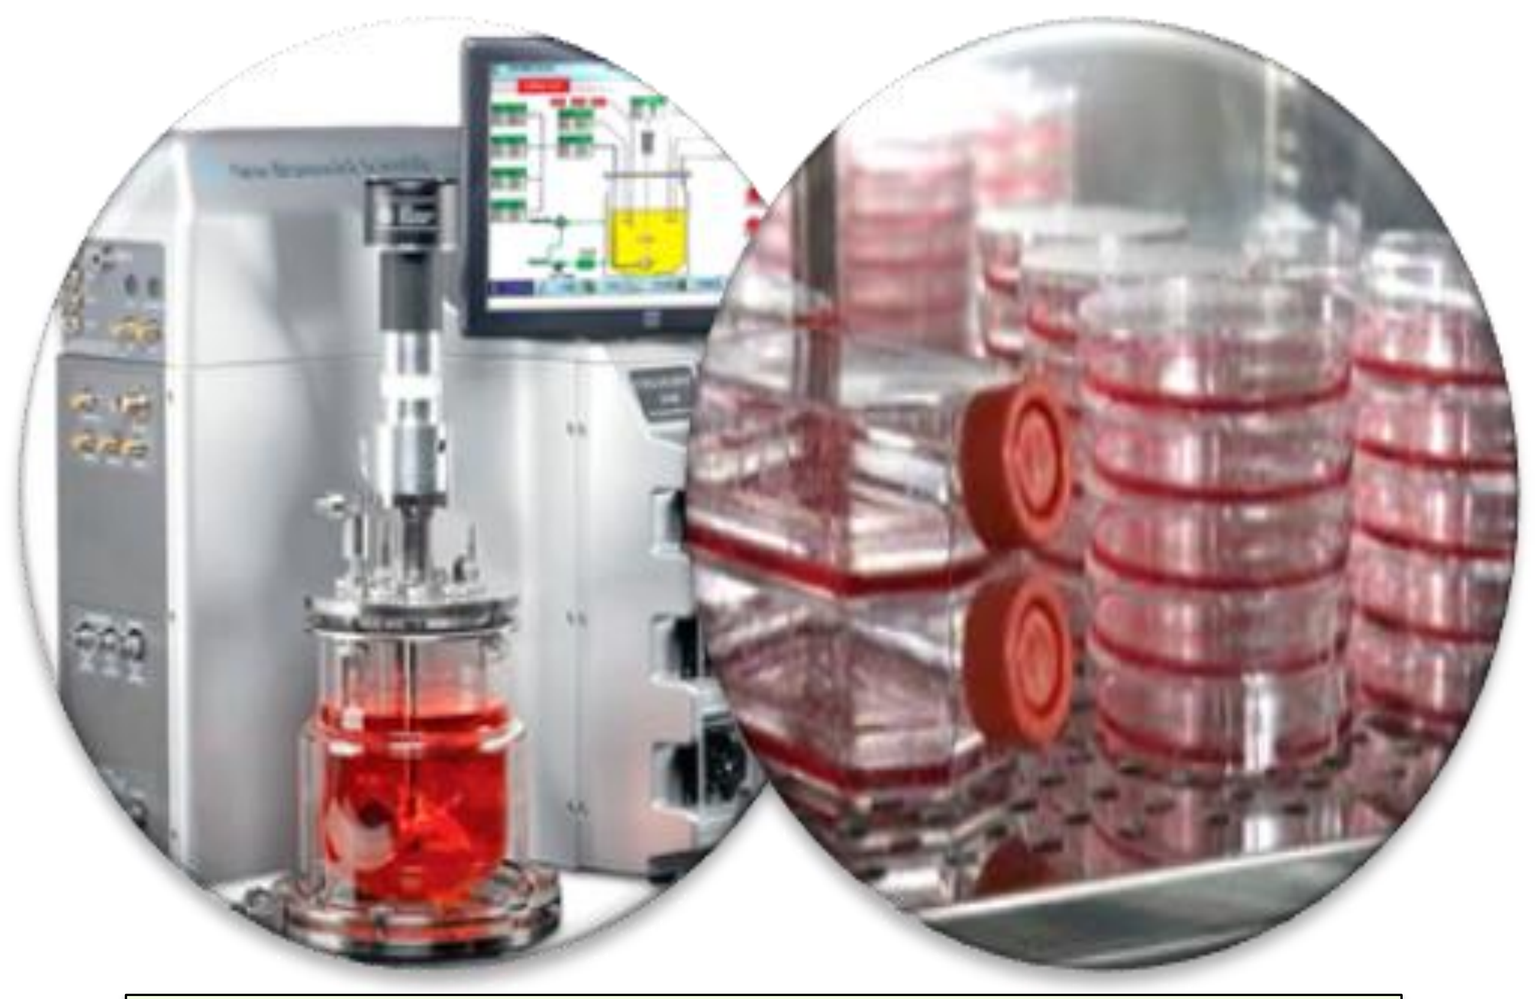

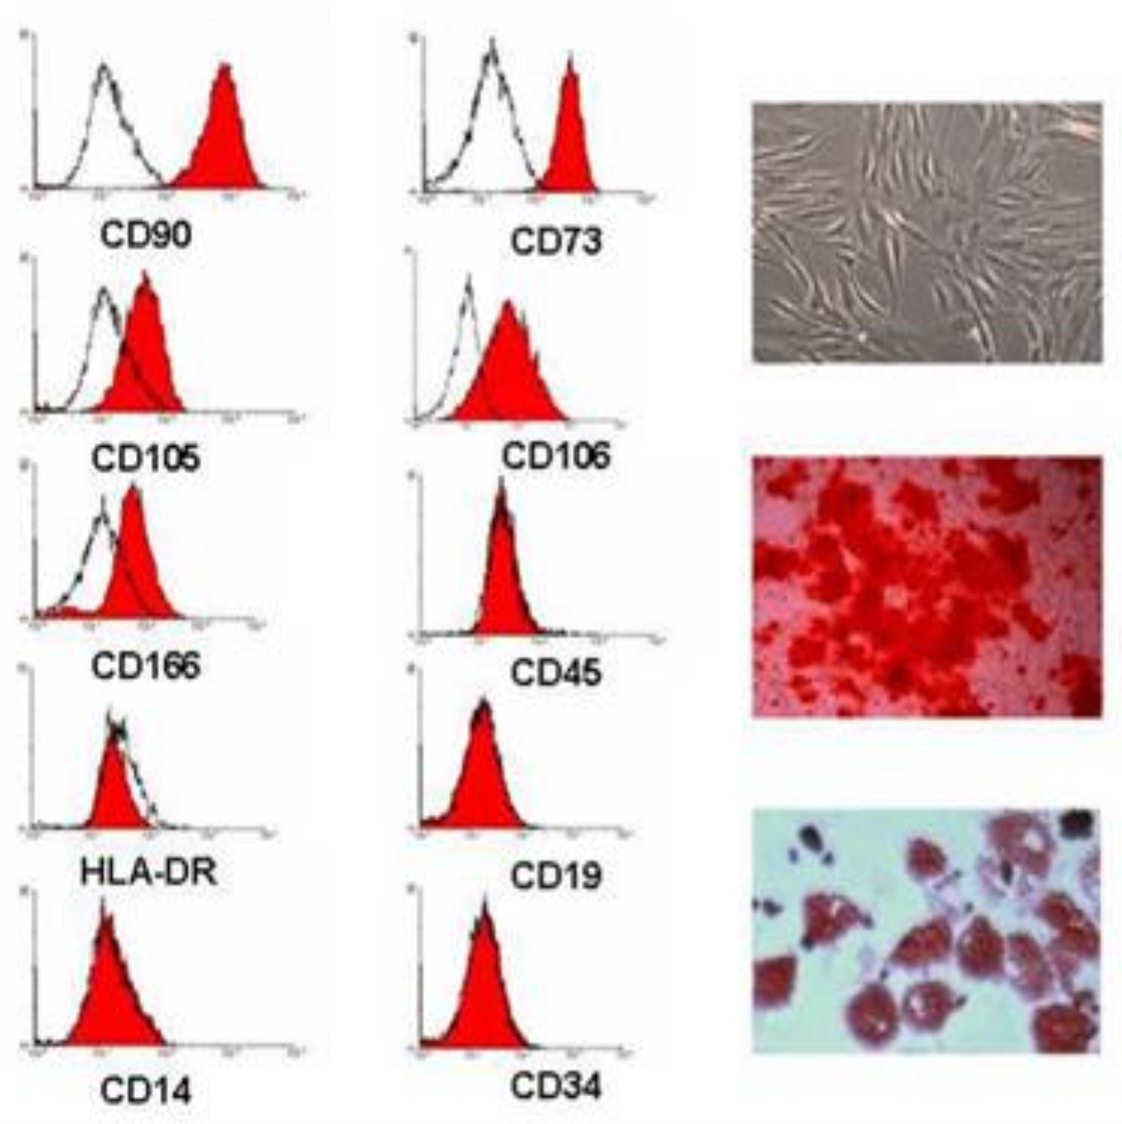
CD90
CD73
CD105
CD106
CD166
CD45
HLA-DR
CD19
CD14
CD34

Flow cytometry for MSC CD45FITC/CD34PE/CD105PerCP/CD73PE/CD90FITC/CD146 and osteogenic differentiation

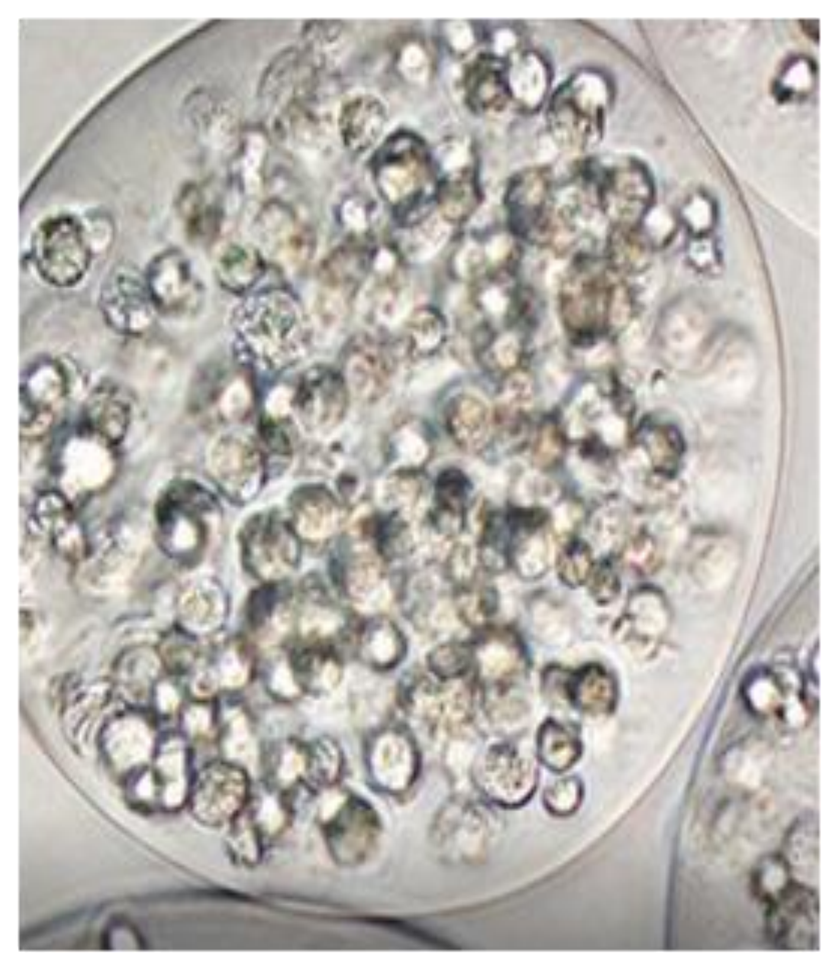

MSC encapsulation

Cell culture with cytokines

SCF, FL, IL3, IL6, DLK-1, TPO

Fold expansion

CD34+ and CXCR4 expression

6 samples of UCB were taken prior to the signing of the informed consent of the parents. Red cell reduction was carried out on the nuclear cell concentrate and then cryopreserved for 1 year until thawed, CD34 + was selection by Kit Miltenyi.

11 umbilical cord samples were obtained to which nucleated cells were obtained by means of vein separation and digestion Wharton Jelly with type II collagenase. The mesenchymal cells were cultured and expanded, which were characterized by immunophenotype CD73 +, CD90 +, CD105 +, CD45 + and CD34 +. The potential for differentiation to osteogenesis was realized with the kit stem cells, the selection of the mesenchymal ones with the CD146 KIT was made and these were expanded.

RESULTS

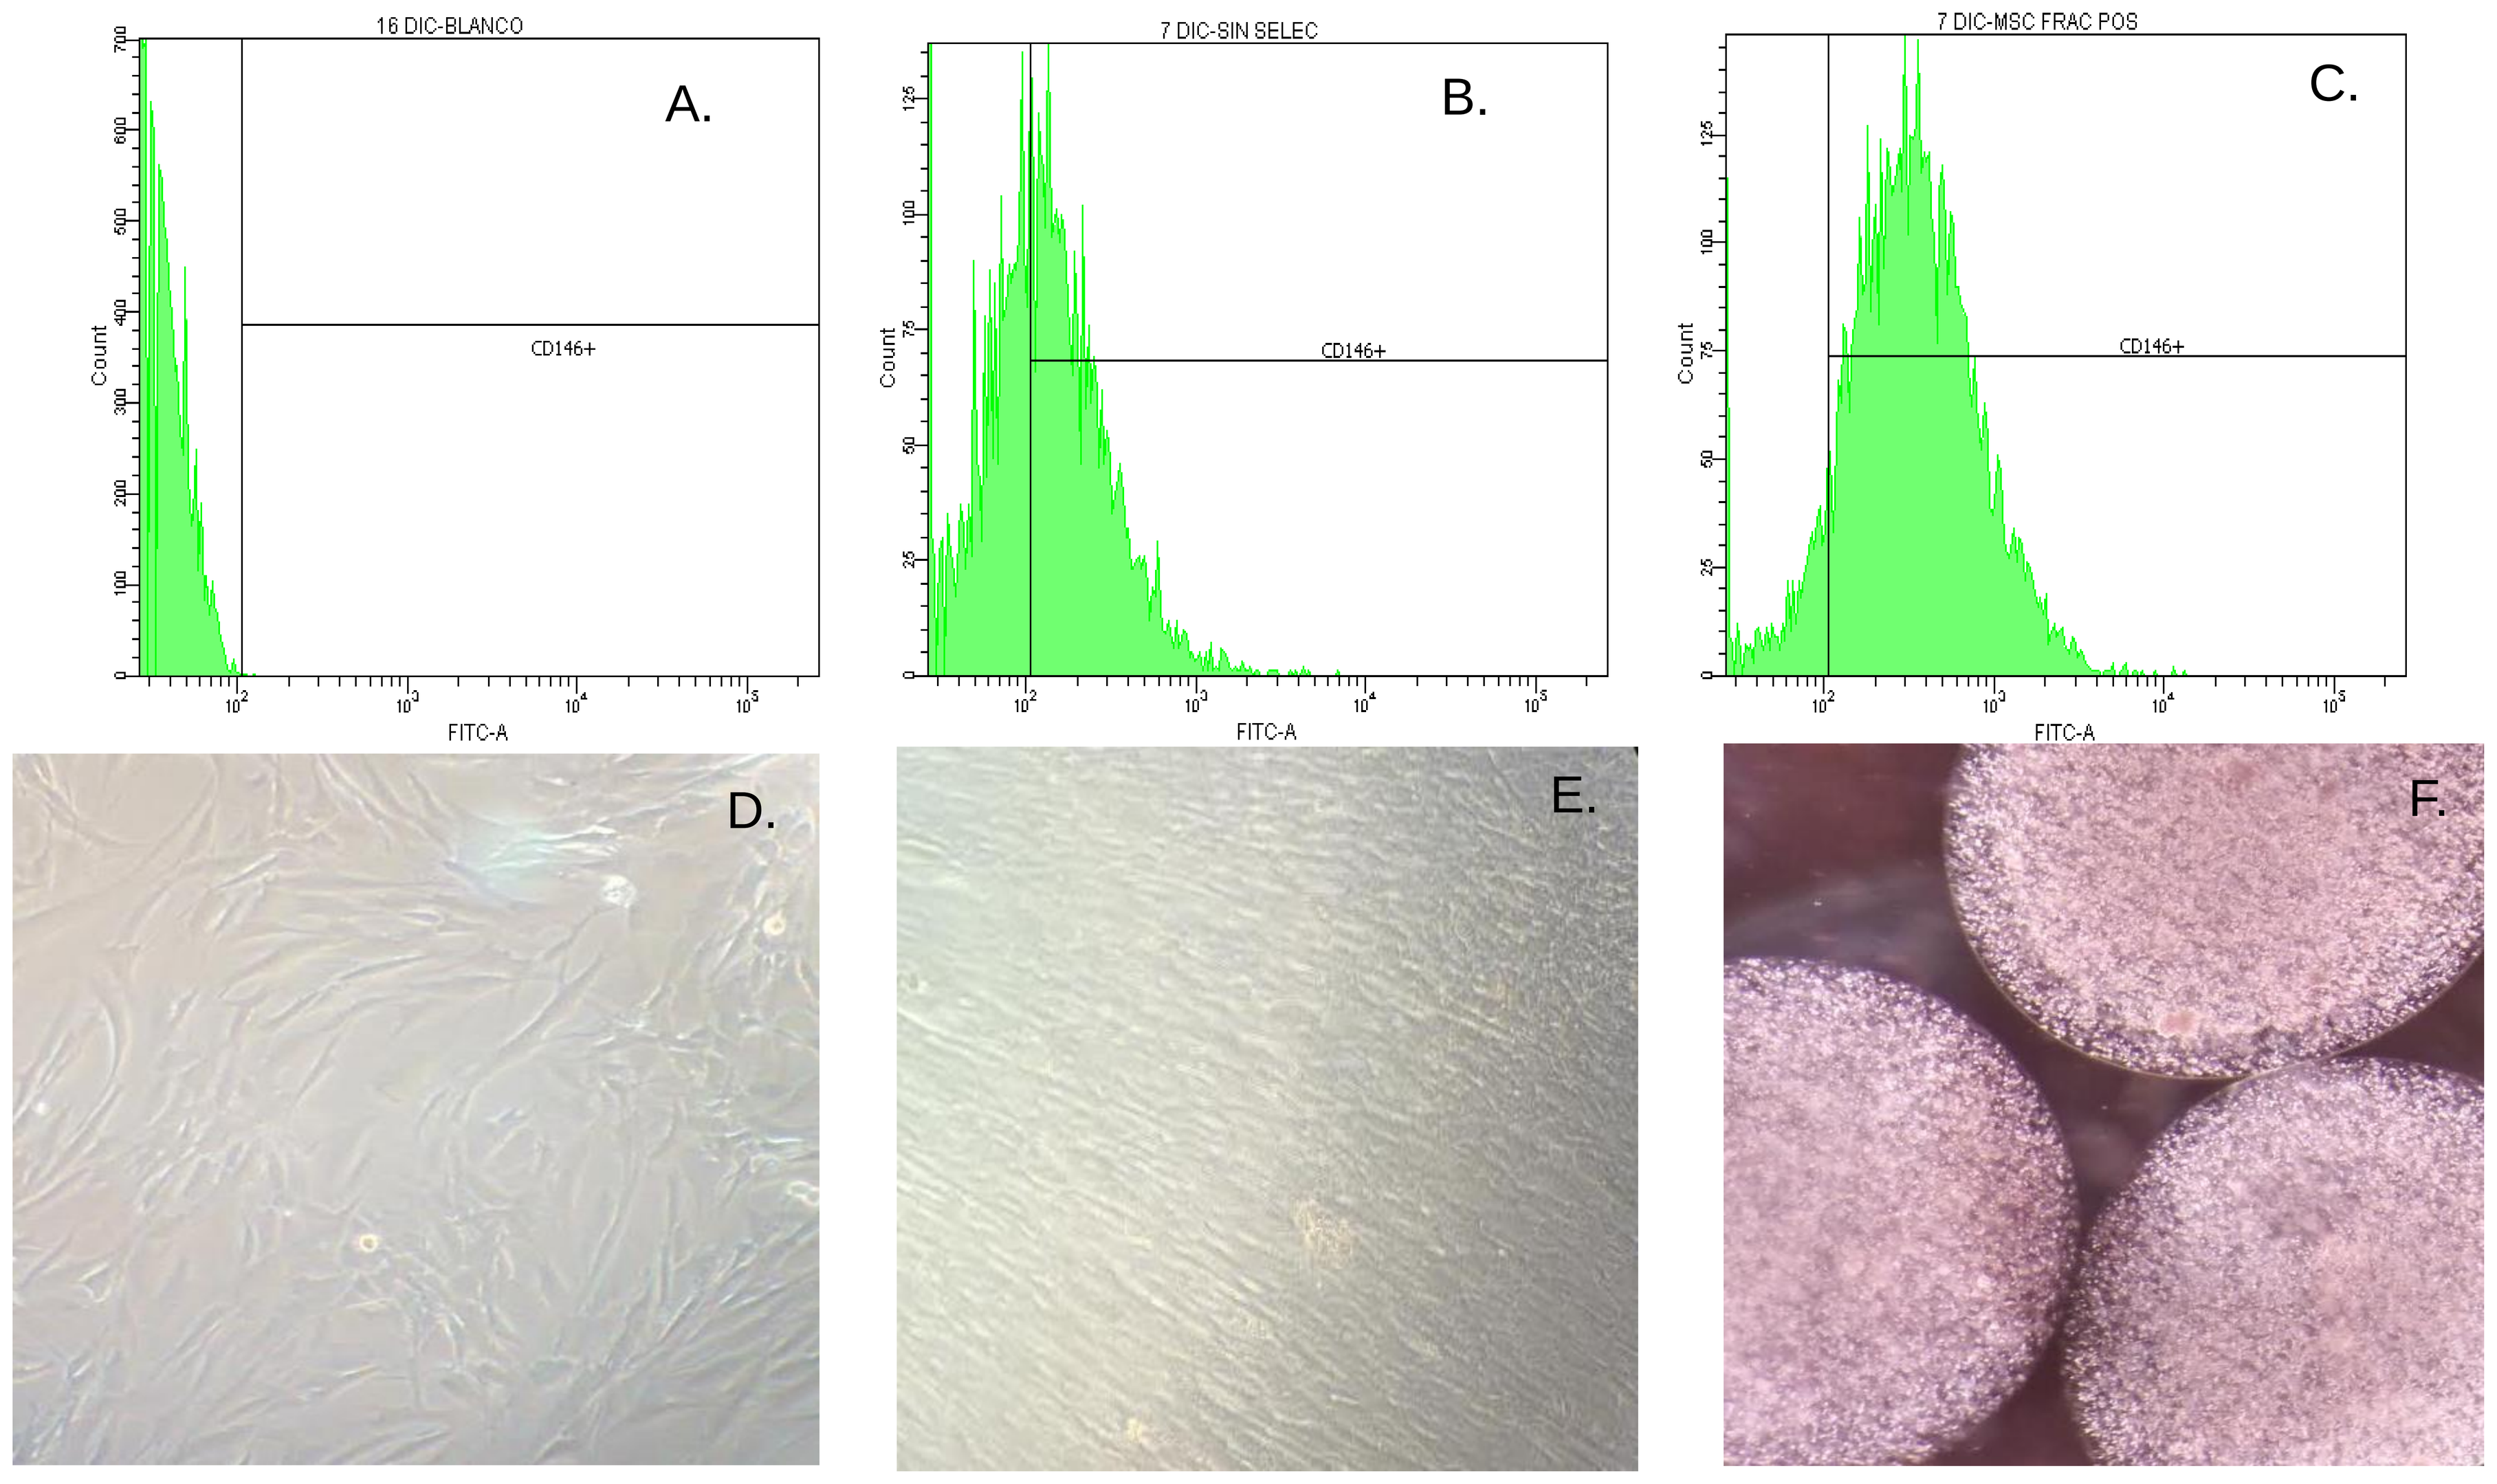
16 DIC-BLANCO
A.
7 DIC-SIN SELEC
B.
7 DIC-MSC FRAC POS
C.
CD146+
Count
FITC-A
D.
E.
F.

Mesenchymal Stem cells from Wharton Jelly. A) White for CD46 +. flow cytometry. B) MSCs WJ pre selection CD146 + 56% C) MSCs WJ post selection CD146 + 90.2%. D and E). MSC confluence 30% and 70% post selection CD146+. F) MSC encapsulated in alginate.

STATICS CULTURE		
NUMBER	CD45/CD34+	CD45/34/49F/90/CXCR4
ASSAY 1	6,23E+00	1,56E+00
ASSAY 2	6,68E+00	1,73E+00
ASSAY 3	6,22E+00	2,39E+00
ASSAY 4	7,50E+00	2,98E+00
ASSAY 5	7,95E+00	2,38E+00
ASSAY 6	6,15E+00	3,17E+00
Average	6,79E+00	2,37E+00

PERFUSIÓN CULTURE		
NUMBER	CD45/CD34+	CD45/34/49F/90/CXCR4
ASSAY 1	1,18E+01	6,61E+00
ASSAY 2	8,54E+00	3,10E+00
ASSAY 3	6,02E+00	2,63E+00
ASSAY 4	1,08E+01	8,69E+00
ASSAY 5	1,08E+01	5,62E+00
ASSAY 6	4,88E+00	4,52E+00
Average	8,80E+00	5,20E+00

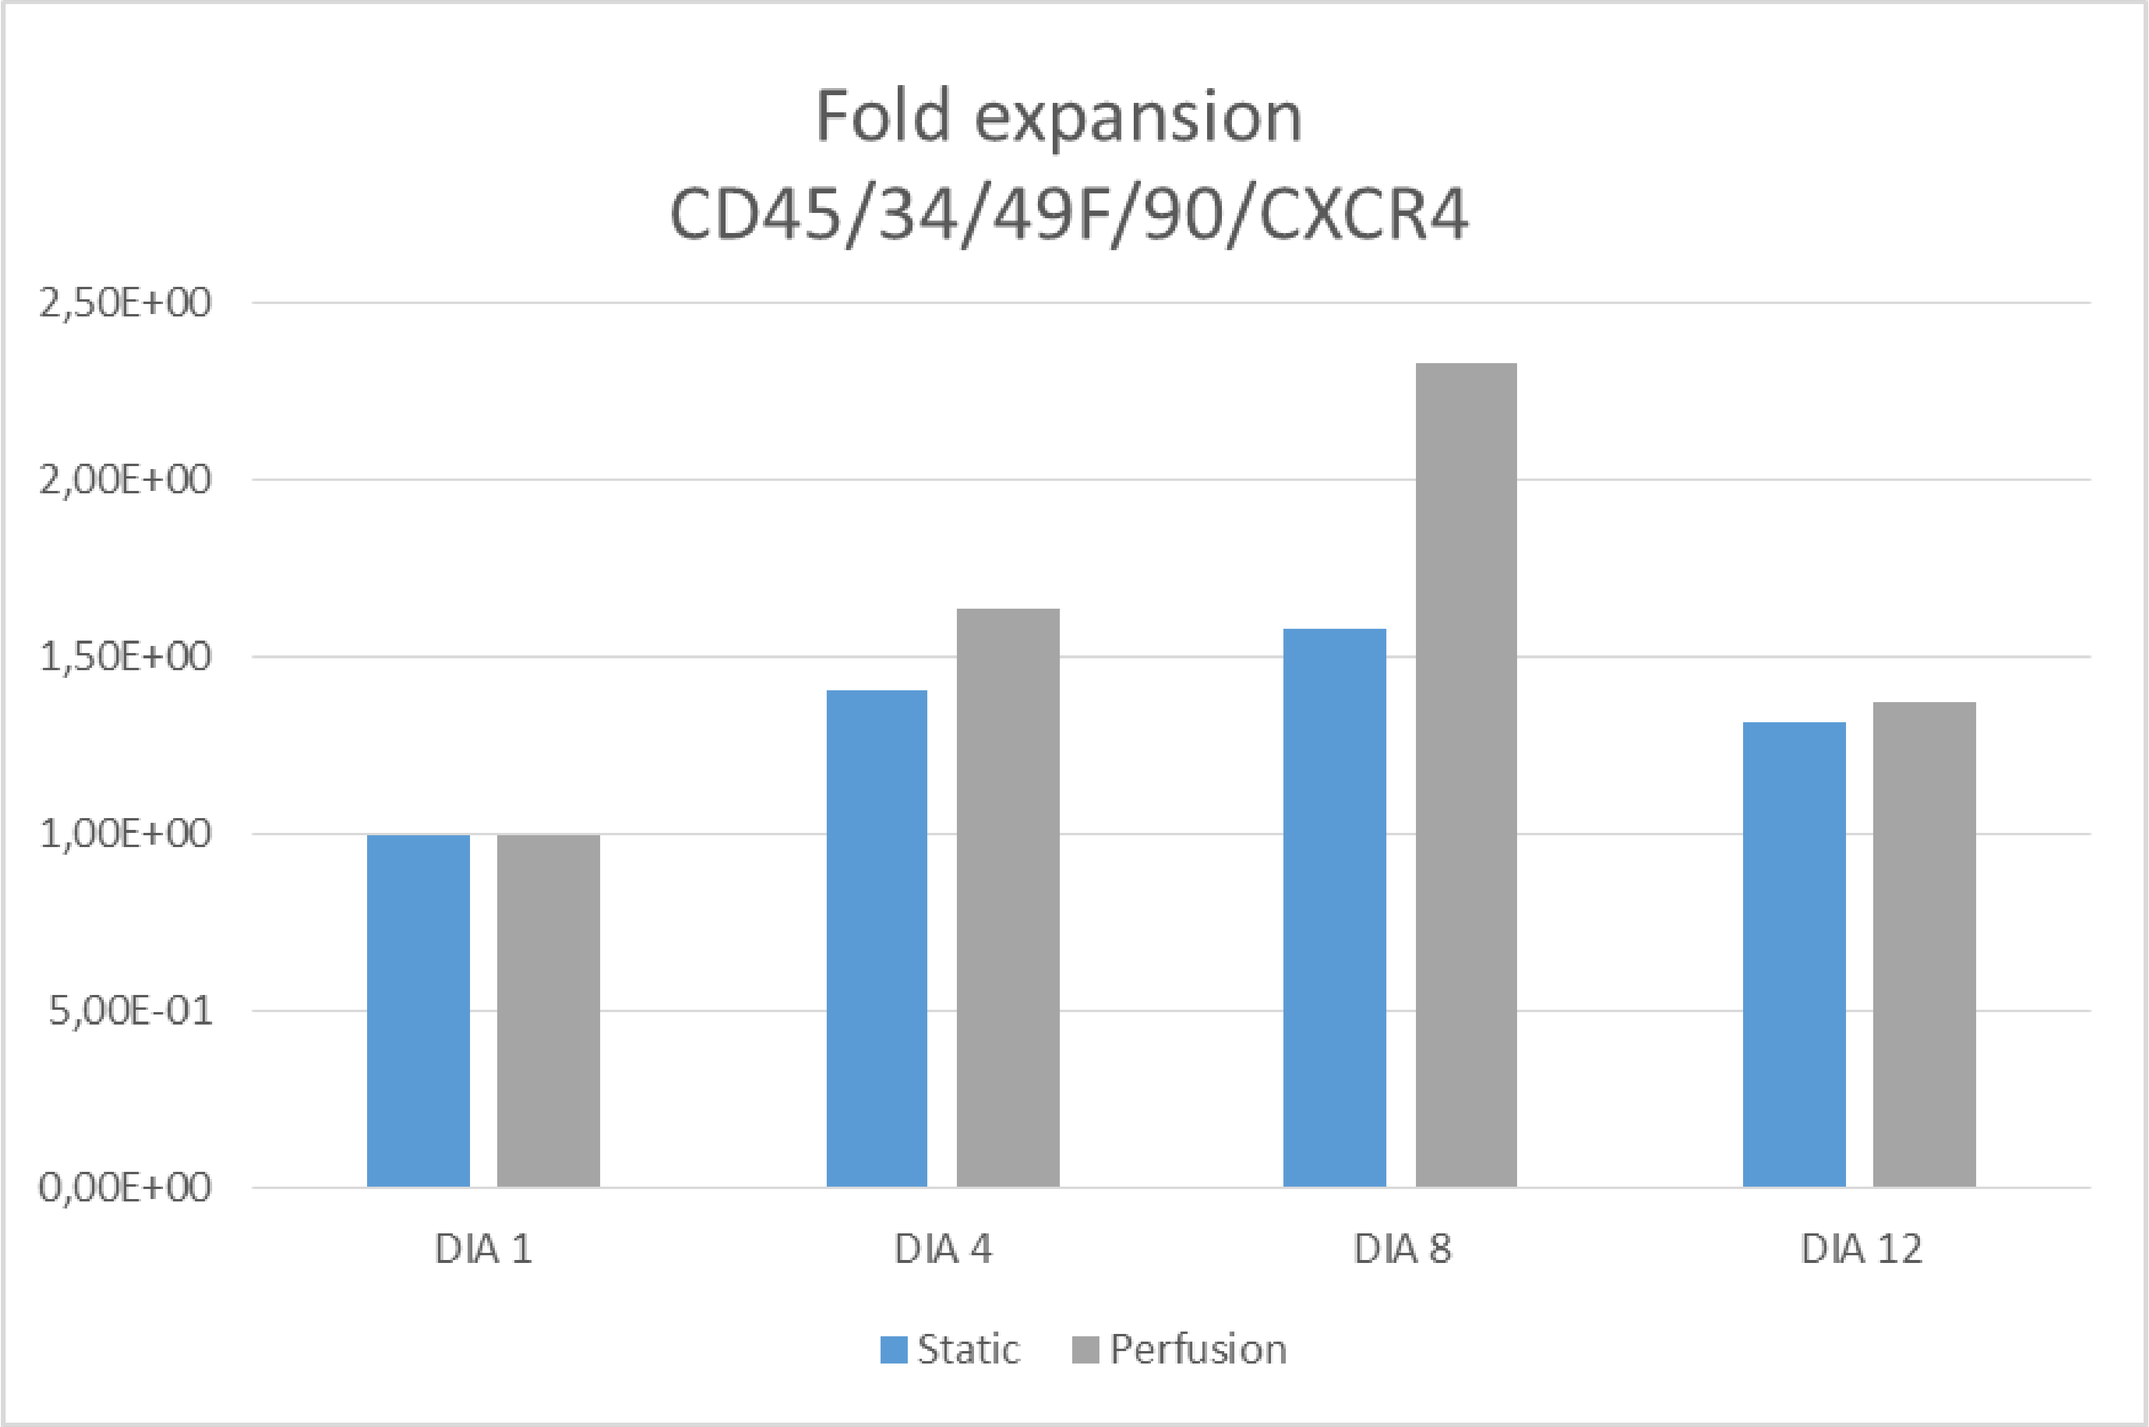
Fold expansion
CD45/34/49F/90/CXCR4
2,50E+00
2,00E+00
1,50E+00
1,00E+00
5,00E-01
0,00E+00
DIA 1
DIA 4
DIA 8
DIA 12
Static
Perfusion

DISCUSION AND CONCLUSIONS

The limitations umbilical cord blood (UCB) transplantation, is the low dosage of haematopoietic stem cell (HSC) and delayed reconstitution, UCB expansion under conventional culture causes rapid cell differentiation of (HSC) responsible for engraftment. In this study, we compared expansión factor of CD34/CD45 ^{DIM} and CD34+/CD90+/CD49F, CXCR4 utilizando culture static Vs Bioreactor, and We employed MSC CD/146+ from Wharton´s Jelly Umbilical Cord encapsulate in alginate calcium beads.

- The model proposed Of perfusion(encapsulated mesenchymal stem cells from Wharton jelly and recombinant human cytokines), is possible increase ex vivo the number of CD34+, CD90+, CD49f cells after 12 days of culture.
- This study evidenced a efficient model of HSC expansion in vivo, however this must be confirming for cultures to long term for confirm the trend shown, also assays in vivo with animal models.
-